DCC-2618, a small molecule inhibitor of normal and mutant KIT kinase for treatment of refractory gastrointestinal stromal tumors (GIST)

Deciphera Pharmaceuticals, Lawrence, KS

INTRODUCTION

Gastrointestinal stromal tumors (GIST) are driven by gain-of-function mutations of the KIT (approx 90%) or PDGFRα (approx 10%) receptor tyrosine kinases. There are over 4,000 new cases of GIST diagnosed each year in the US. Current targeted kinase inhibitor therapy with imatinib and sunitinib is initially successful but resistance mutations develop over a period of 1 or more years. Using Deciphera Pharmaceutical's approach to kinase inhibition, DCC-2618 has been designed to effectively inhibit the imatinib and sunitinib-sensitive KIT juxtamembrane domain mutants (JMD) as well as secondary resistant KIT kinase-domain mutants. DCC-2618 additionally targets PDGFRα oncogenic mutants. The data demonstrate that Deciphera's technology has identified DCC-2618 as a molecule which can be developed as second line therapy for imatinib and sunitinib resistant GIST patients with the potential to progress to frontline GIST therapy. This presentation will highlight the attributes of DCC-2618 which is in preclinical development for treatment of refractory GIST.

KEY ATTRIBUTES OF KINASE SWITCH POCKET INHIBITORS

-Switch pockets are a previously unexplored "druggable" region of kinases discovered at Deciphera.
-These pockets are part of the machinery kinases use to regulate their activities under normal conditions. However, mutation of these switch control mechanisms leads to uncontrolled kinase activity and cancer.
-This approach to kinase regulation is complementary to more classical ATP pocket inhibitors (i.e. sunitinib).
-This approach retains potency vs. gatekeeper and other mutant forms of oncogenic kinases.
-Provide an approach for developing highly selective kinase inhibitors or inhibitors with unique multi-kinase inhibitory profiles
-Switch Pocket kinase inhibitors exhibit resiliency to high cellular ATP concentrations known to exist in cancer cells.
-DCC-2618 acts by imposing an inactive conformation (shape) of highly resistant and aggressive secondary mutations of KIT kinase.

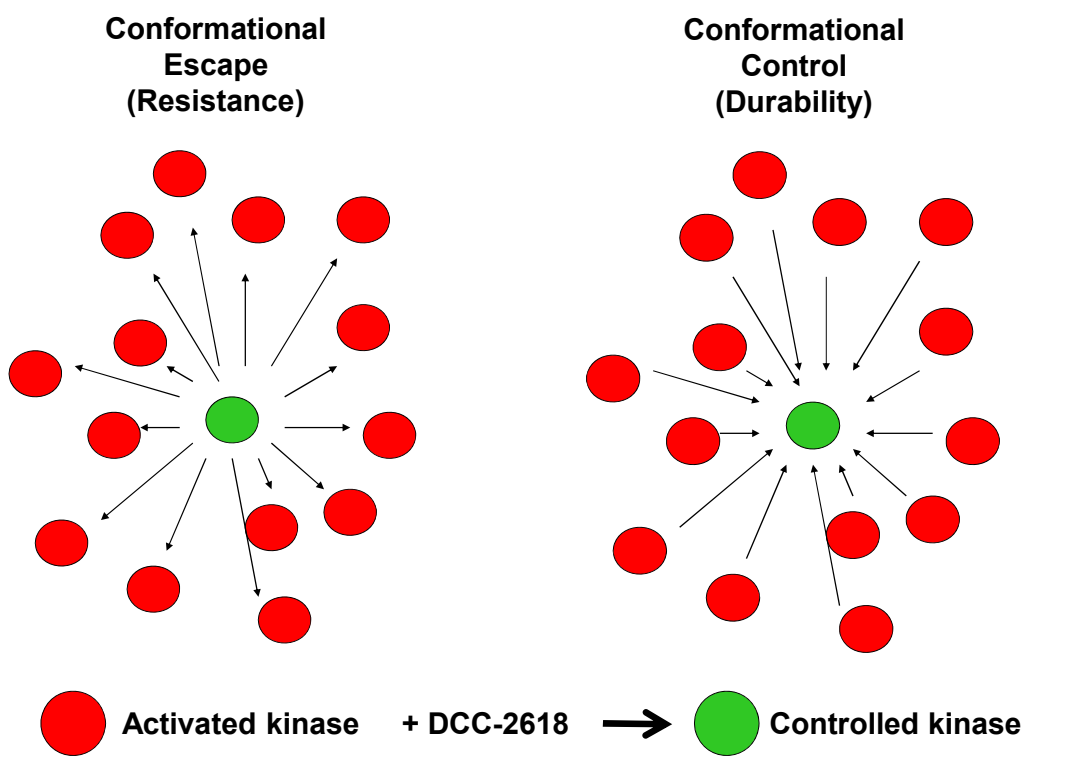

Conformational
Escape
(Resistance)
Conformational
Control
(Durability)
Activated kinase
+ DCC-2618
Controlled kinase

DCC-2618 remains potent over time

JMD inhibitory domain (exon 11) primary mutation and D816V (exon 17) is secondary mutation					
IC_{50} (nM)	1-2 h	2-3 h	4-5 h	6-7 h	8-9 h
Sunitinib	0.240	0.820	0.920	0.860	0.790
DCC-2618	0.011	0.012	0.009	0.015	0.009

RESULTS

DCC-2618 inhibits normal and mutant KIT kinase

	KIT Mutant Profile, IC_{50} (nM)					
	wt c-KIT	KIT V654A exon 13	KIT T670I exon 14	KIT D816H exon 17	KIT D816V exon 17	KIT JMD ΔVV/D816V exons 11&17
Imatinib	27	339	3,500	>10,000	> 5,000	>10,000
Sorafenib	10	156	358	481	>10,000	2,900
Sunitinib	5	110	10	1,800	2,200	768
DCC-2618	6	9	18	5	14	9

DCC-2618 inhibits other cancer related kinases

	Solid Tumor Kinases, IC_{50} (nM)			
	PDGFR-α	PDGFR-β	KDR	cFMS
Sorafenib	1.4	9	6	1.4
Sunitinib	162	29	86	14
DCC-2618	30	13	17	10
	Vascular Maturation/ Stabilization		Neo- Vascularization	Bone Mets

DCC-2618 profile vs 300 human kinases

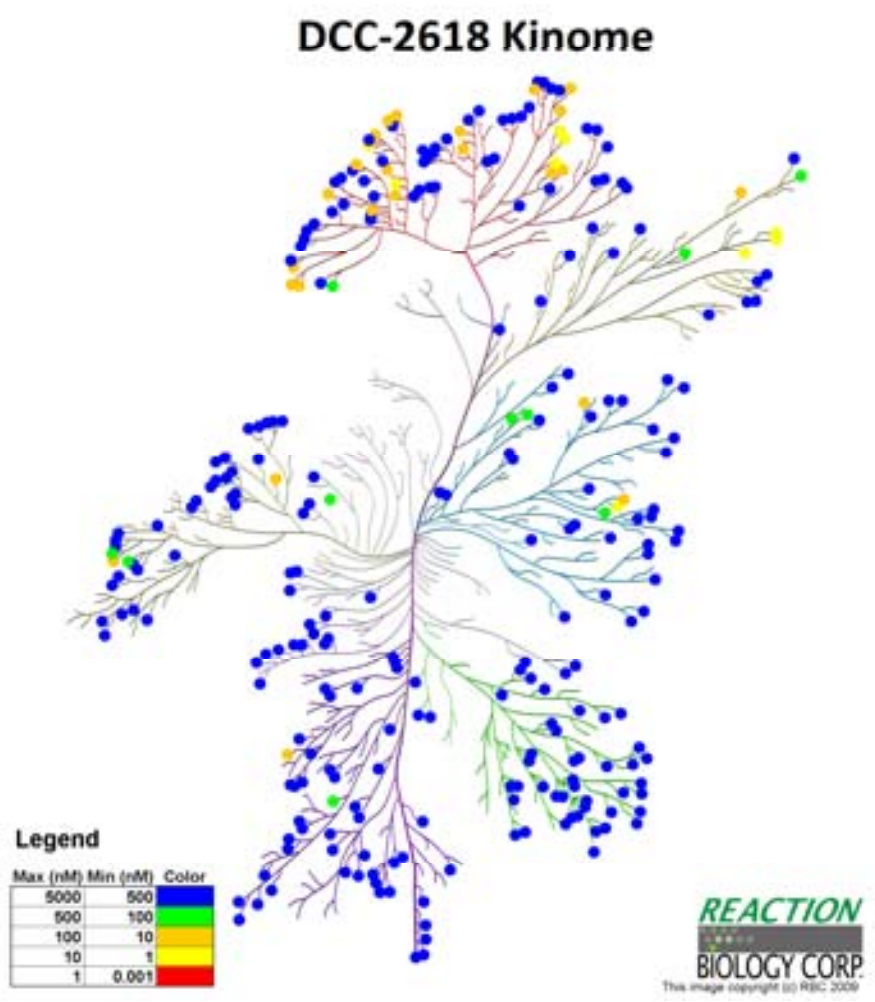

DCC-2618 Kinome
Legend
Max (nM) Min (nM) Color
5000 500
500 100
100 10
10 1
1 0.001
REACTION BIOLOGY CORP.

DCC-2618 inhibits resistant Exon 17 KIT mutations in cells

IC_{50} (nM)	CHO pKIT western Ex 17 D816V	CHO pKIT western Ex 11 ΔVV Ex 17 D816V	CHO pKIT western Ex 17 D820A
Sorafenib	2,600	2,600	53
Imatinib	>7,500	>5,000	>7,500
Sunitinib	>7,500	>5,000	1,900
DCC-2618	106	43	67

DCC-2618 inhibits mutant KIT in GIST patient cell lines

IC_{50} (nM)	GIST T1 pKIT western Ex 11 deletion	GIST 430 pKIT western (ΔJM/V654A) Ex 11/Ex 14	GIST 48 pKIT western (V560D/D820A) Ex 11/Ex 17
Sorafenib	5	162	489
Imatinib	12	>5,000	>5,000
Sunitinib	3	37	5,000
DCC-2618	2	5	32

DCC-2618 inhibits KIT in GIST tumor xenografts after single dose

50 mpk DCC-2618

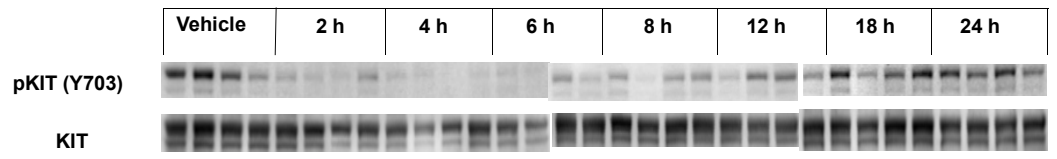

Vehicle
2 h
4 h
6 h
8 h
12 h
18 h
24 h
pKIT (Y703)
KIT

	2 hr	4 hr	6 hr	8 hr	12 hr	18 hr	24 hr
% Inhibition	79%	88%	74%	69%	39%	8%	0%

25 mpk DCC-2618

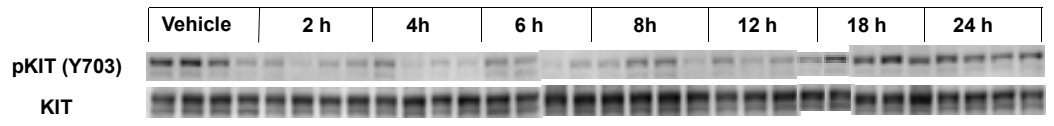

Vehicle
2 h
4h
6 h
8h
12 h
18 h
24 h
pKIT (Y703)
KIT

	2 hr	4 hr	6 hr	8 hr	12 hr	18 hr	24 hr
% Inhibition	64%	66%	62%	58%	54%	10%	16%

SUMMARY

-DCC-2618 is a kinase switch inhibitor that can control drug resistant mutants of KIT and PDGFRα in GIST

-Current therapies are unable to address all escape mutants

-DCC-2618 is orally bioavailable and is currently in preclinical development for refractory GIST

-A Phase I trial with refractory GIST patients is planned for Q4 2011

www.deciphera.com